Impact of Heat-Stable Factor VIII Mimetics and Replacement Therapies on Inhibitor Titer Using a Modified Nijmegen-Bethesda Test Across Bovine Chromogenic FVIII Assays

Abstract Number: PB2087

Tara Quinton, Karen M. Black, Ali Sadeghi-Khomami
Precision BioLogic Inc., Dartmouth, Nova Scotia, Canada

Presented at ISTH 2026
July 11–15, 2026

PrecisionBioLogic

Introduction

Accurate measurement of factor VIII (FVIII) inhibitors in hemophilia A (HA) is increasingly complex due to the use of FVIII mimetics and recombinant FVIII (rFVIII) replacement therapies. Humanized bispecific antibodies such as emicizumab and Mim8 interfere with conventional clot-based FVIII assays, while rFVIII products differ in their susceptibility to heat inactivation, a critical step in modified Nijmegen-Bethesda inhibitor testing.

Altuviiio® (Altuvoct®), an extended half-life rFVIII, exhibits greater resistance to heat inactivation[1] compared with conventional rFVIII replacement products such as Advate®[2] The combined presence of FVIII mimetics, FVIII inhibitors, and rFVIII replacement therapies poses analytical challenges for reliable inhibitor measurement in clinical laboratories.

Our aim was to evaluate the impact of FVIII mimetics and rFVIII replacement therapies with differing heat stability on FVIII inhibitor measurement using a modified Nijmegen-Bethesda assay combined with bovine chromogenic FVIII methods.

Methods

Patient inhibitor plasma (George King Bio-Medical) was adjusted to target concentrations of 0.2, 1, 5, and 10 Bethesda units (BU/mL) using pooled congenital severe-HA plasma (HRF) and Factor VIII polyclonal antibody (Affinity Biologicals).

Samples were supplemented with Mim8 (0, 5, 8, 10, and 20 µg/mL), emicizumab (0, 25, 50, 75, and 150 µg/mL), or rFVIII products (Altuviiio and Advate) at 0.0, 0.05, 0.2, 0.8, and 1.0 IU/mL and stored frozen at ≤-70 °C.

FVIII inhibitor activity was measured in triplicate using CRYO*check*™ Factor VIII Inhibitor Kit incorporating heat inactivation (56 °C for 30, 60, 90 or 120 min) followed by centrifugation at 2700 × *g* for 5 min, together with bovine CRYO*check* Chromogenic Factor VIII (Precision BioLogic) on an IL ACL TOP 700 CTS.

Selected samples were also evaluated by two alternative chromogenic assays (Coatest® SP4 FVIII (Chromogenix) on an IL ACL TOP 550 and Factor VIII Chromogenic Assay (Siemens) on a BCS XP).

Results

FVIII inhibitor titers measured using the modified Nijmegen-Bethesda assay with bovine chromogenic FVIII showed consistent recovery across all Mim8 and emicizumab concentrations and across the full inhibitor range of 0.2–10 BU/mL **(Figures 1 and 2)**.

Superior inactivation of heat-sensitive Advate was observed regardless of the duration of heat inactivation **(Figure 3)**. In contrast, samples containing Altuviiio demonstrated increased residual FVIII activity that was reduced by prolonged heat inactivation.

Titer measurements decreased with increasing rFVIII concentrations, particularly at lower inhibitor levels with a misclassification risk **(Figure 4)**.

Conclusions

Bovine chromogenic FVIII assays are not affected by Mim8 or emicizumab interference and can be used in FVIII inhibitor testing. Comparable trends were observed between the new bovine CRYO*check* Chromogenic Factor VIII and both the Coatest and Siemens assays.

Heat-inactivation in FVIII inhibitor measurement refines the accuracy of borderline titers in samples with low FVIII levels.[3] However, low titer 1.0 BU/mL plasma samples containing ≥0.20 IU/mL of Advate or Altuviiio could still be misclassified as inhibitor negative (<0.6 BU/mL), despite achieving a negligible <1% FVIII activity after a prolonged and effective heat inactivation step. This might be due to the engagement of inhibitor antibodies with excess FVIII denatured molecules. Our results have yet to be verified in clinical inhibitor plasma samples with specific antibodies against each rFVIII molecule.

References

1. Bowyer A. *Haemophilia*. 2026, 32(1):306–308.
2. Payne AB et al. *Haemophilia*. 2020, 26(1):e28–e30.
3. Miller CH et al. J Thromb Haemost. 2012 Jun;10(6):1055–61.

Figure 1

Factor VIII inhibitor titer of plasma spiked with emicizumab at 0 to 150 µg/mL.

The measured titer (BU/mL; n=3 replicates) of inhibitor negative plasma (0.2 BU/mL) or inhibitor positive plasma (1.0, 5.0 and 10.0 BU/mL target levels) spiked with 0, 25, 50, 75 and 150 µg/mL of emicizumab.

Testing was performed using CRYO*check* Factor VIII Inhibitor Kit in conjunction with CRYO*check* Chromogenic Factor VIII (bovine), Coatest SP4 FVIII and Siemens Factor VIII Chromogenic Assay.

The threshold for a positive titer (≥0.6 BU/mL) is indicated by the blue dotted line for the 0.2 BU/mL plasma. The acceptance limits (±25% of the un-spiked sample results for each assay) are indicated by the red dotted lines for inhibitor positive samples.

*The therapeutic level of emicizumab is approximately 50 µg/mL.

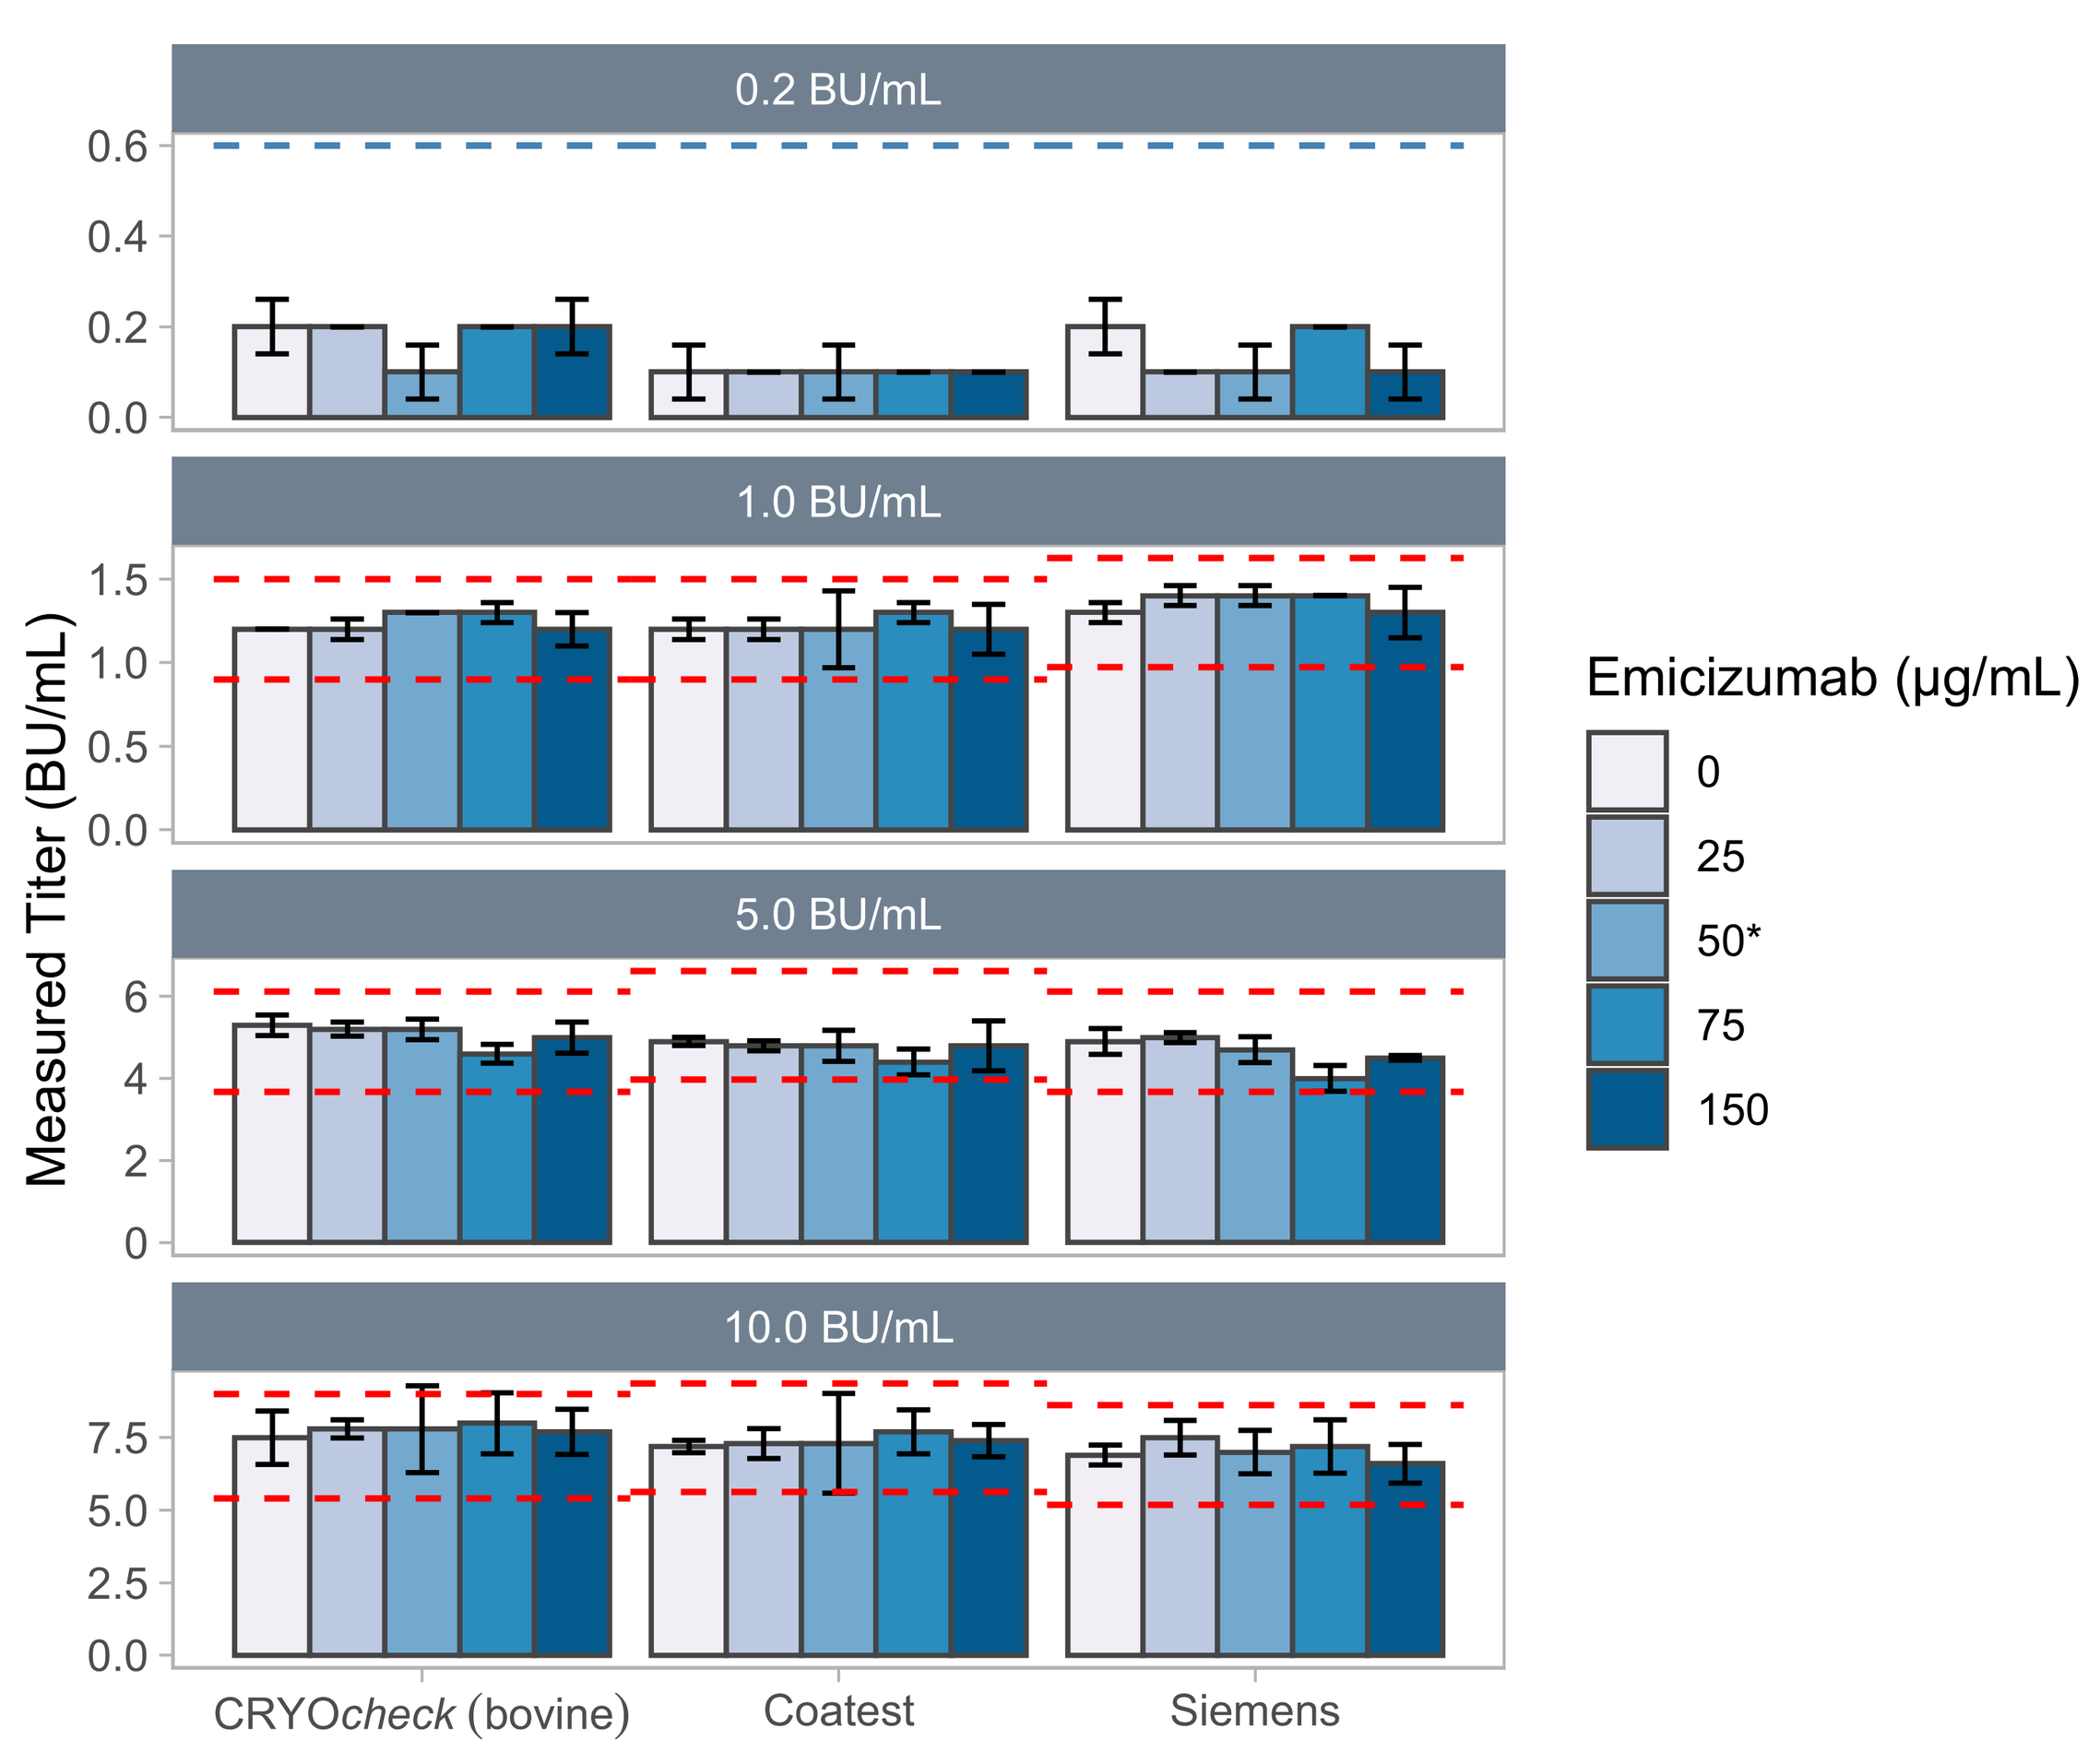
0.2 BU/mL
1.0 BU/mL
5.0 BU/mL
10.0 BU/mL
Measured Titer (BU/mL)
Emicizumab (µg/mL)
0
25
50*
75
150
CRYOcheck (bovine)
Coatest
Siemens

Figure 2

Factor VIII inhibitor titer of plasma spiked with Mim8 at 0 to 20 µg/mL.

The measured titer (BU/mL; n=3 replicates) of inhibitor negative plasma (0.2 BU/mL) or inhibitor positive plasma (1.0, 5.0 and 10.0 BU/mL target levels) spiked with 0, 5, 8, 10 and 20 µg/mL of Mim8.

Testing was performed using CRYO*check* Factor VIII Inhibitor Kit in conjunction with CRYO*check* Chromogenic Factor VIII (bovine), Coatest SP4 FVIII and Siemens Factor VIII Chromogenic Assay.

The threshold for a positive titer (≥0.6 BU/mL) is indicated by the blue dotted line for the 0.2 BU/mL plasma. The acceptance limits (±25% of the un-spiked sample results for each assay) are indicated by the red dotted lines for inhibitor positive samples.

*The expected therapeutic level of Mim8 is 5 µg/mL.

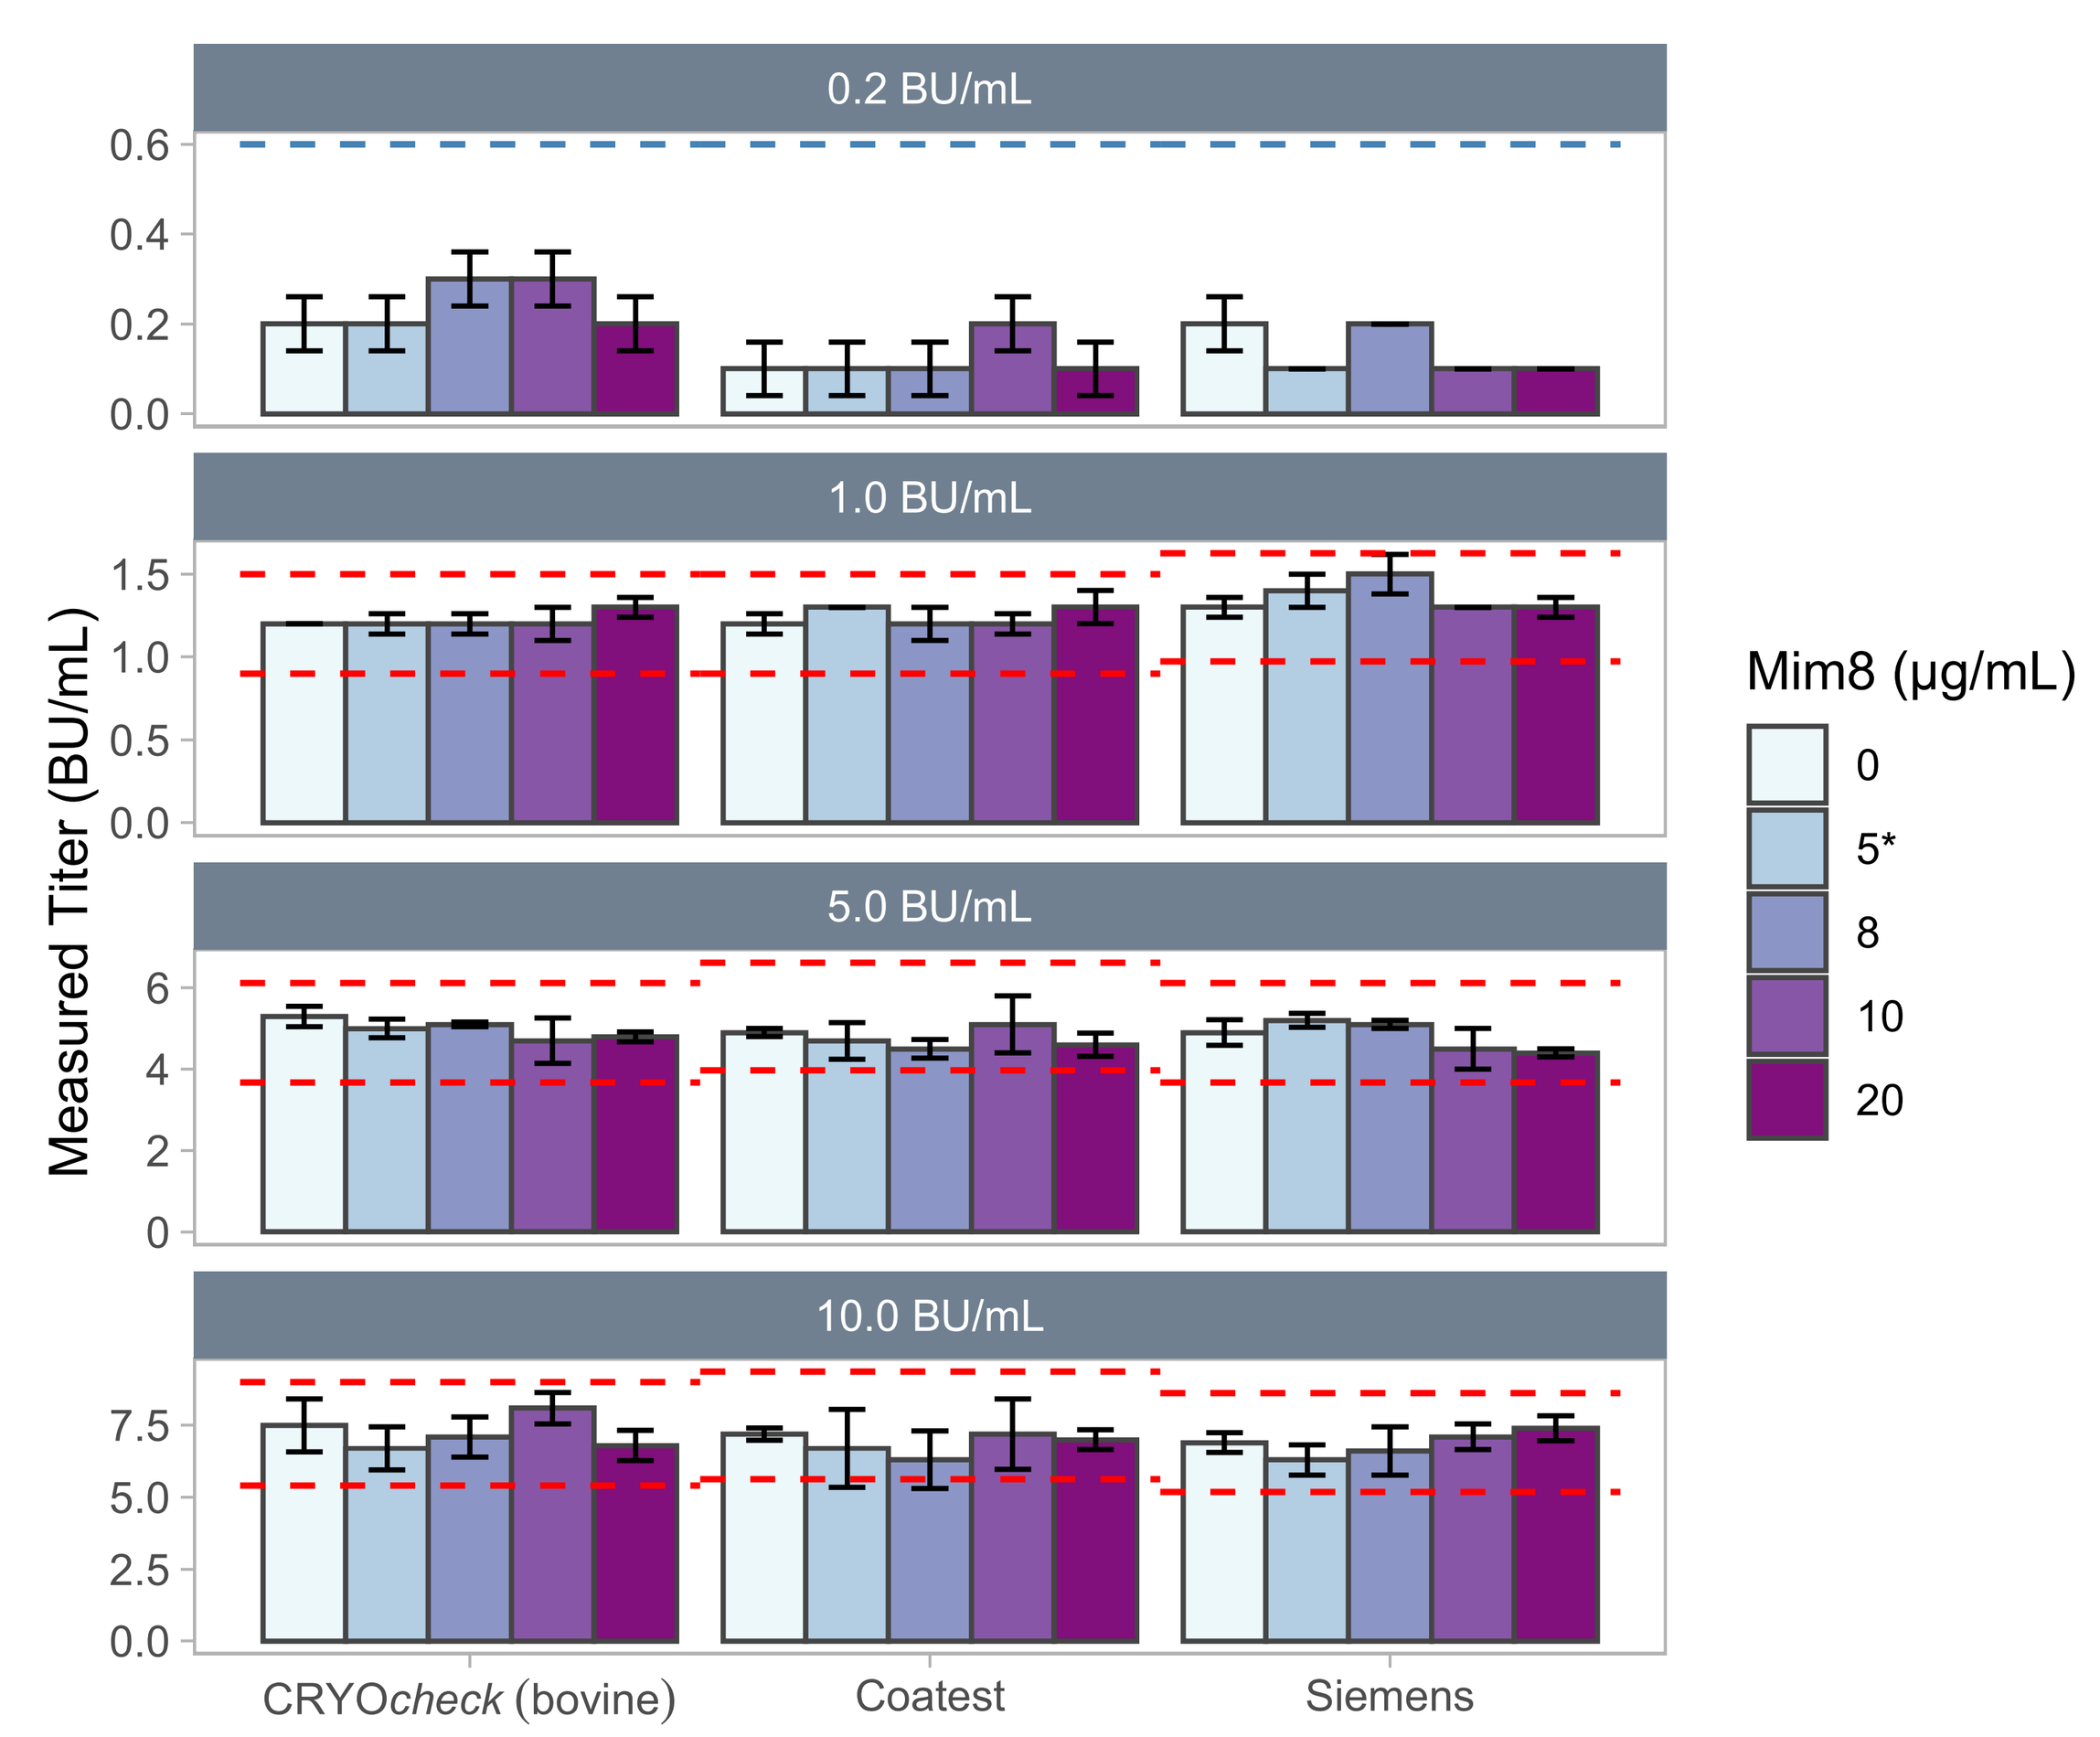
0.2 BU/mL
1.0 BU/mL
5.0 BU/mL
10.0 BU/mL
Measured Titer (BU/mL)
Mim8 (µg/mL)
0
5*
8
10
20
CRYOcheck (bovine)
Coatest
Siemens

Figure 3

Factor VIII activity of deficient plasma spiked with Advate or Altuviiio at 0.05, 0.2, 0.8 or 1.0 IU/mL after heat inactivation.

The measured factor VIII activity (%; n=3 replicates) of congenital FVIII deficient plasma spiked with 0.05, 0.2, 0.8 or 1.0 IU/mL of Advate or Altuviiio after four different heat inactivations (30 min, 60 min, 90 min and 120 min at 56 °C).

Testing was performed using CRYO*check* Chromogenic Factor VIII (bovine).

Although chromogenic assays overestimate Alutviiio approximately 2.5-fold, a conversion factor was not applied.

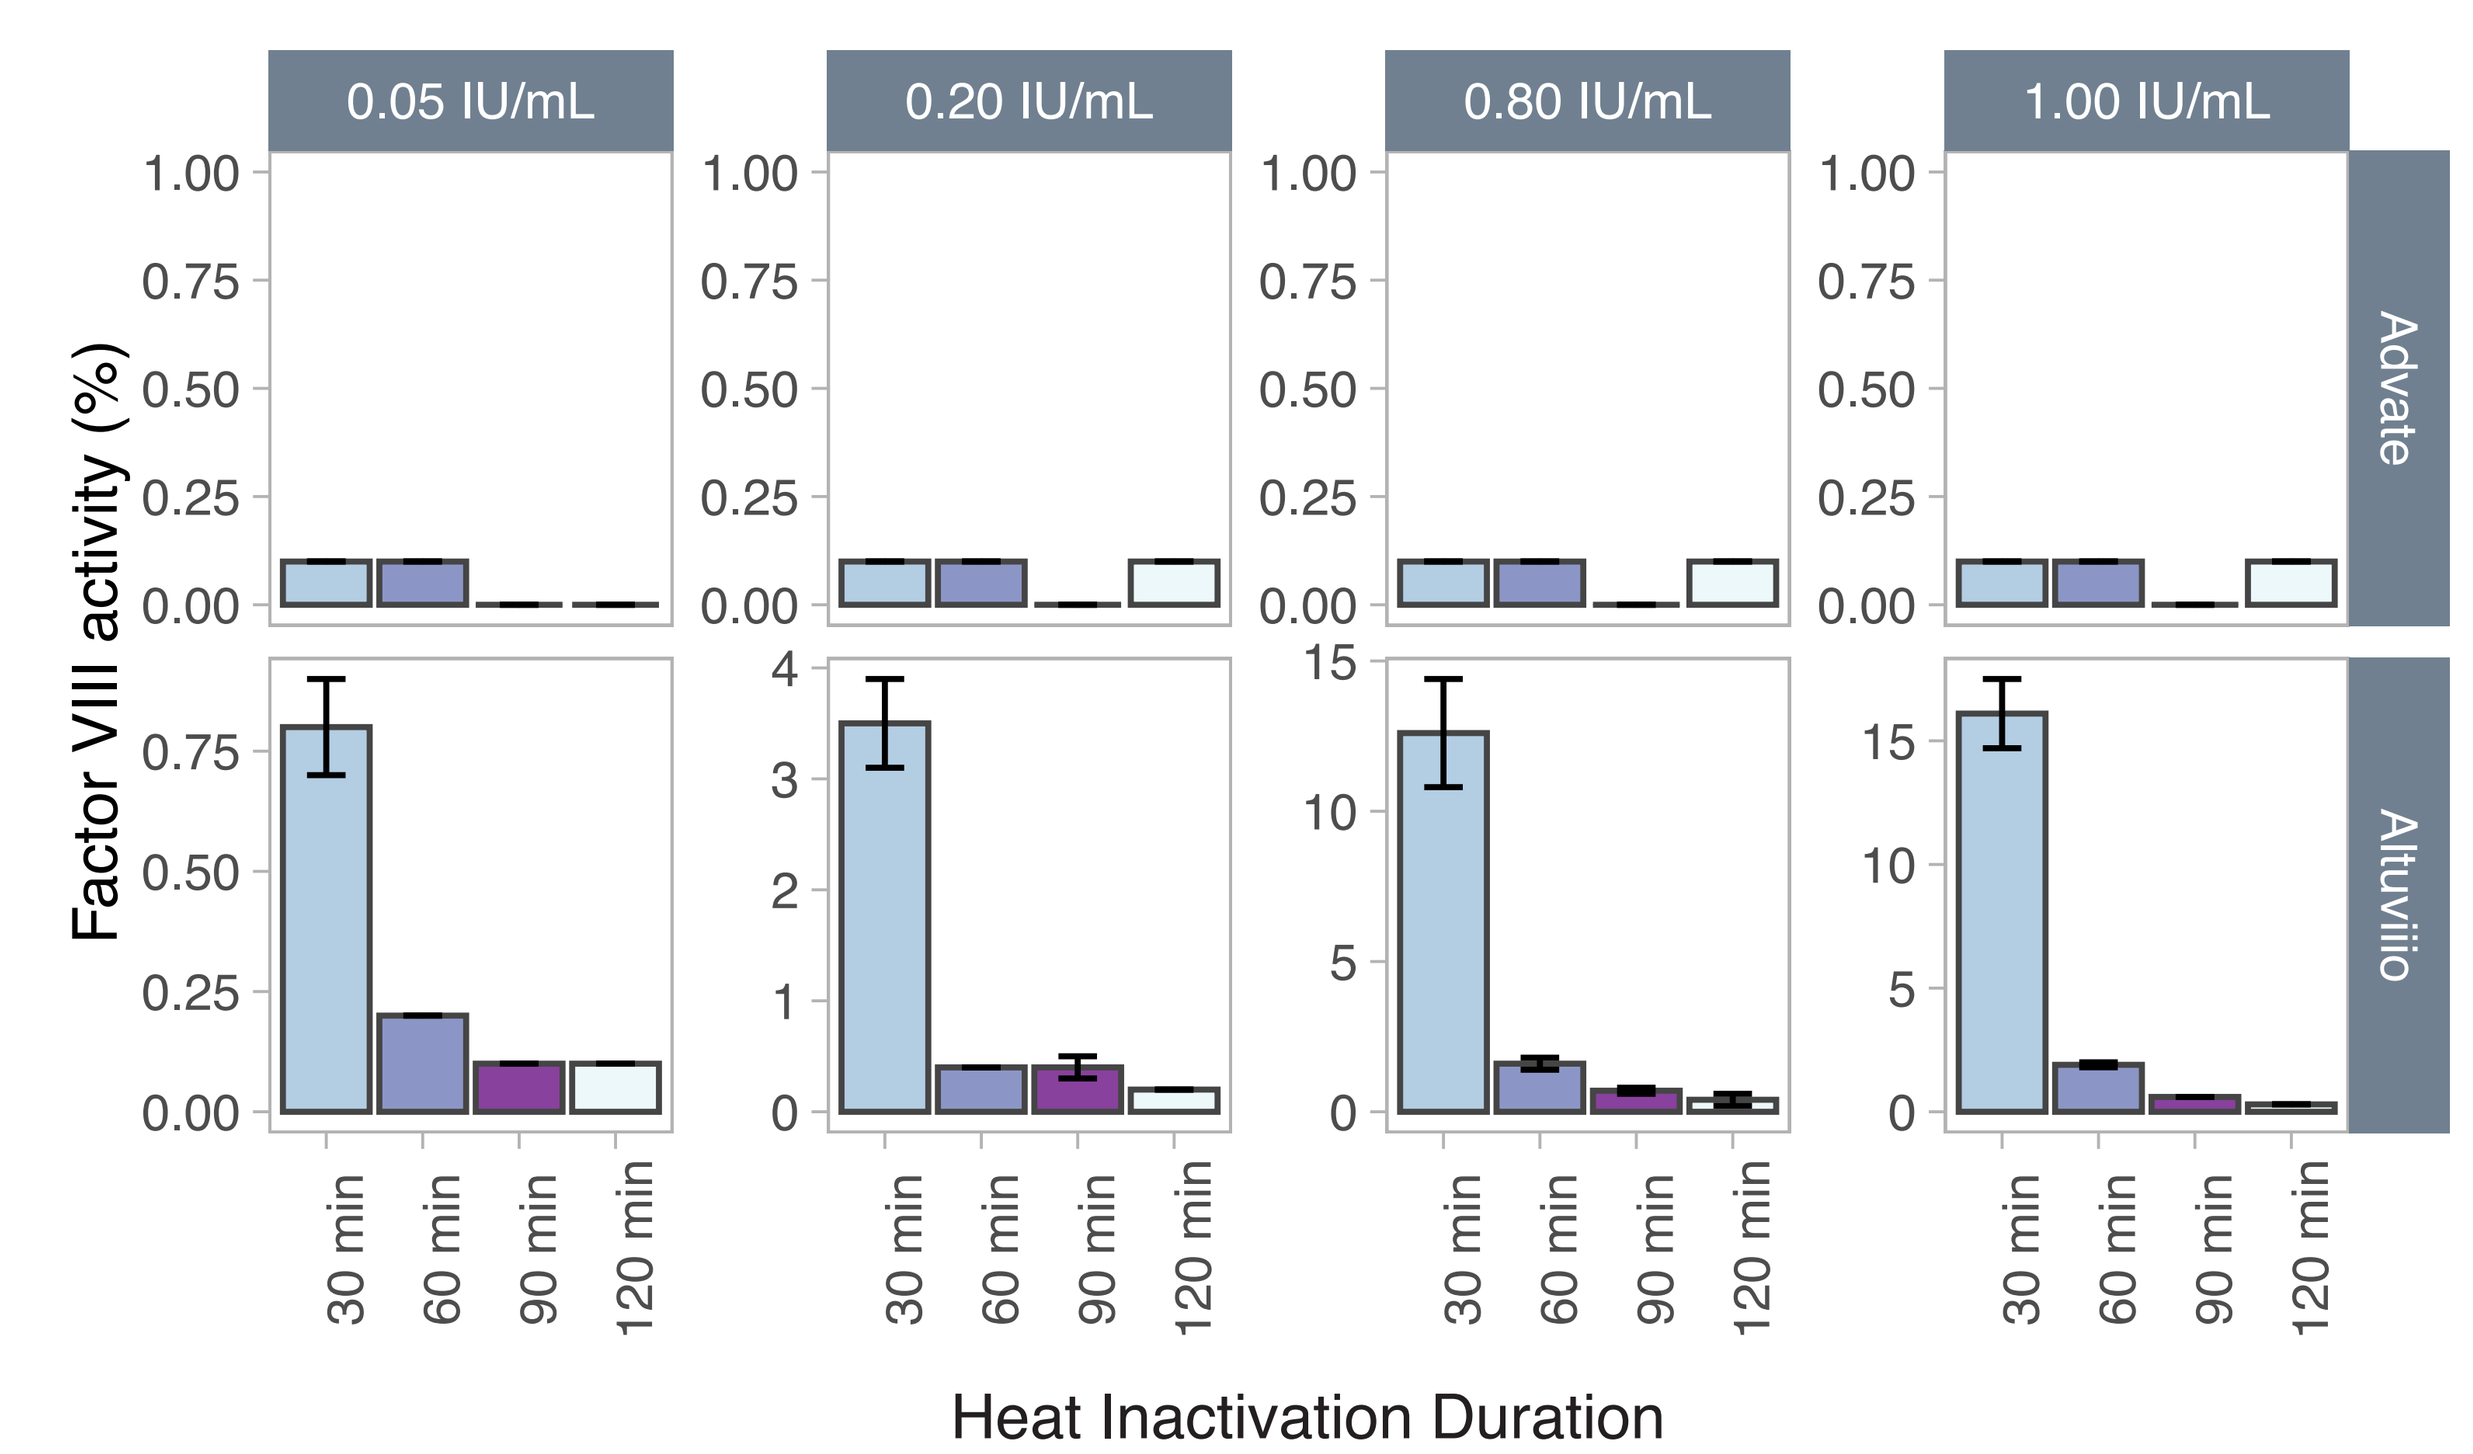
0.05 IU/mL
0.20 IU/mL
0.80 IU/mL
1.00 IU/mL
Advate
Altuviiio
Factor VIII activity (%)
30 min
60 min
90 min
120 min
Heat Inactivation Duration

Figure 4

Factor VIII inhibitor titer of 1.0 BU/mL plasma spiked with Advate or Altuviiio at 0.0, 0.05, 0.2, 0.8 or 1.0 IU/mL after heat inactivation.

The measured titer (BU/mL; n=3 replicates) of inhibitor positive plasma (1.0 BU/mL) spiked with 0.0, 0.05, 0.2, 0.8 or 1.0 IU/mL of Advate or Altuviiio after four different heat inactivations (30 min, 60 min, 90 min and 120 min at 56 °C).

Testing was performed using CRYO*check* Factor VIII Inhibitor Kit in conjunction with CRYO*check* Chromogenic Factor VIII (bovine).

The threshold for a positive titer (≥0.6 BU/mL) is indicated by the blue dotted line. The measured titer of the unspiked (0.0 IU/mL) inhibitor positive plasma after each heat inactivation is indicated by the red dotted line segments.

Similar outcomes were observed with and without centrifugation (5 min at 2700 × *g*) following heat inactivation.

Data for 0.2, 5.0 and 10.0 BU/mL target levels is not shown but a similar under-recovery of titer was observed at ≥0.2 IU/mL rFVIII.

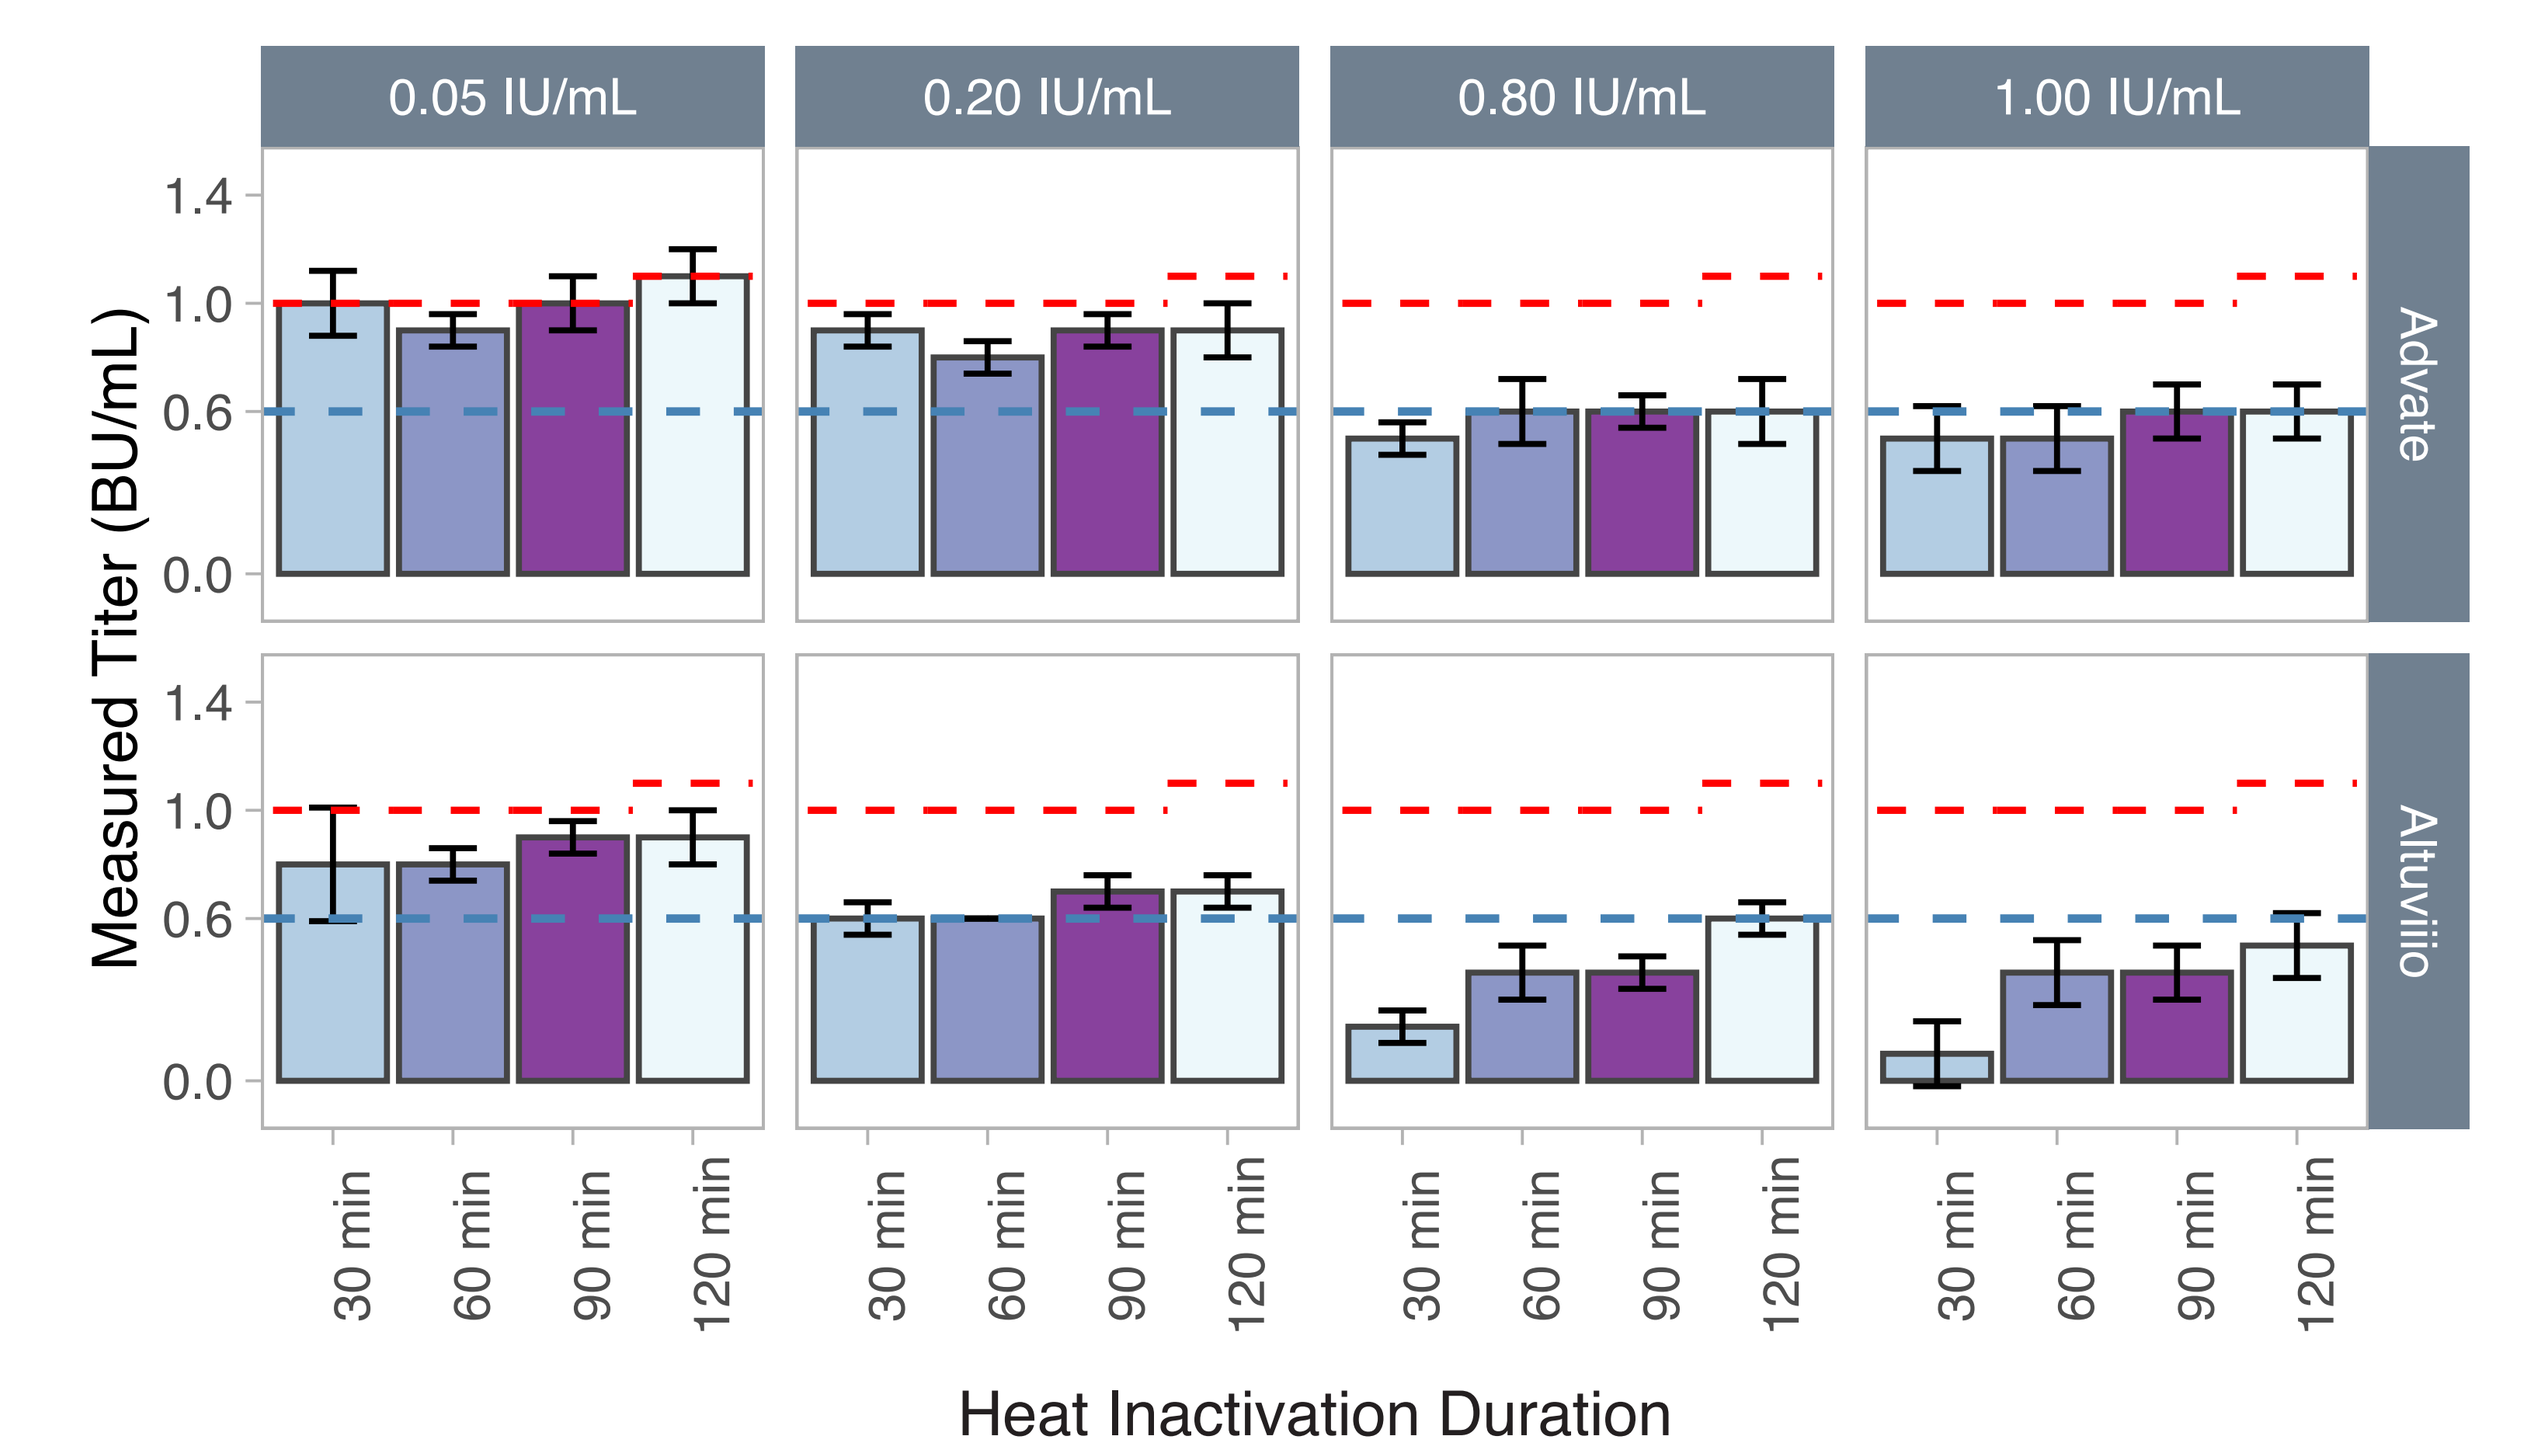
0.05 IU/mL
0.20 IU/mL
0.80 IU/mL
1.00 IU/mL
Advate
Altuviiio
Measured Titer (BU/mL)
30 min
60 min
90 min
120 min
Heat Inactivation Duration